Poster No. 805

CROI 2015

Helen Kovari
Division of Infectious Diseases
and Hospital Epidemiology
University Hospital Zurich
University of Zurich
Rämistrasse 100
8091 Zurich, Switzerland
+41 44 255 91 95
helen.kovari@usz.ch

D:A:D

Antiretroviral Drugs Associated with Chronic ALT Elevations in Persons without HCV and HBV Infection

H Kovari[1], C Sabin[2], B Ledergerber[1], L Ryom[3], A d'Arminio Monforte[4], M Law[5], S De Wit[6], C Pradier[7] , F Dabis[8] , P Reiss[9], A Phillips[2], JD Lundgren[3], R Weber[1] and the D:A:D Study Group

[1]Division of Infectious Diseases and Hospital Epidemiology, University Hospital Zurich, Switzerland; [2]Research Department of Infection and Population Health, UCL, United Kingdom; [3]CHIP, Department of Infectious Diseases and Rheumatology, Section 2100, Rigshospitalet – University of Copenhagen, Denmark; [4]Hospital San Paolo, University of Milan, Italy; [5]Kirby Institute, University of New South Wales, Sydney, NSW, Australia; [6]Department of Infectious Diseases, St Pierre University Hospital, Brussels, Belgium, [7]Department of Public Health, Nice University Hospital, Nice, France, [8]Centre Inserm U0897-Epidémiologie-Biostatistique, Bordeaux, France, [9]Div. of Infectious Diseases and Dept. of Global Health, Academic Medical Center, University of Amsterdam, Amsterdam, Netherlands

BACKGROUND

- HIV-positive persons frequently have chronic elevated liver enzymes, even without HCV- or HBV-coinfection [1].
- Most studies have focused on severe liver enzyme elevations (LEE), defined as 3-5 times the upper limit of normal or more [2], but only limited data are available on elevations just above normal limits. The underlying cause of chronic LEE is often unclear.

Study Aim

- To identify risk factors associated with chronic alanine aminotransferase (ALT) elevation above normal limits, including demographic, clinical and HIV-specific variables, focusing on antiretroviral therapy (ART).

METHODS

- The D:A:D Study is a prospective cohort collaboration of >49,000 HIV-positive persons from 11 cohorts in Europe, Australia, and the United States.
- Chronic LEE for males/females was defined as ALT >50/>35 U/L at ≥2 visits spanning at least 6 months within 2 years.
- ART exposure was categorized as follows: no exposure; ongoing exposure for < or ≥2 years after initiation; and discontinuation for < or ≥2 years.
- D:A:D participants without HCV/HBV infection, with ≥3 ALT measurements, ≥6 months of follow-up and normal ALT at baseline were followed from study entry to the earliest of chronic LEE, death, 1st February 2013, or last follow-up. Cohort specific baseline dates were chosen according to the introduction of routine ALT monitoring.
- Poisson regression was used to assess chronic LEE and its' association with ART and traditional risk factors.

RESULTS

- A total of 18,060 persons without HCV/HBV and with normal ALT at baseline were included in the analyses (**Figure 1**).
- Over 92,059 person-years (PY), 5412 participants developed chronic LEE (incidence 5.88/100 PY [95% CI 5.72-6.04]).
- Baseline characteristics are displayed in **Table 1**. Median number of ALT measurements per person was 14 (IQR 8-23). APRI score was >1.5 in 5% and FIB-4 Score was >3.25 in 6% of 10,470 persons with available scores (58%).

Figure 1

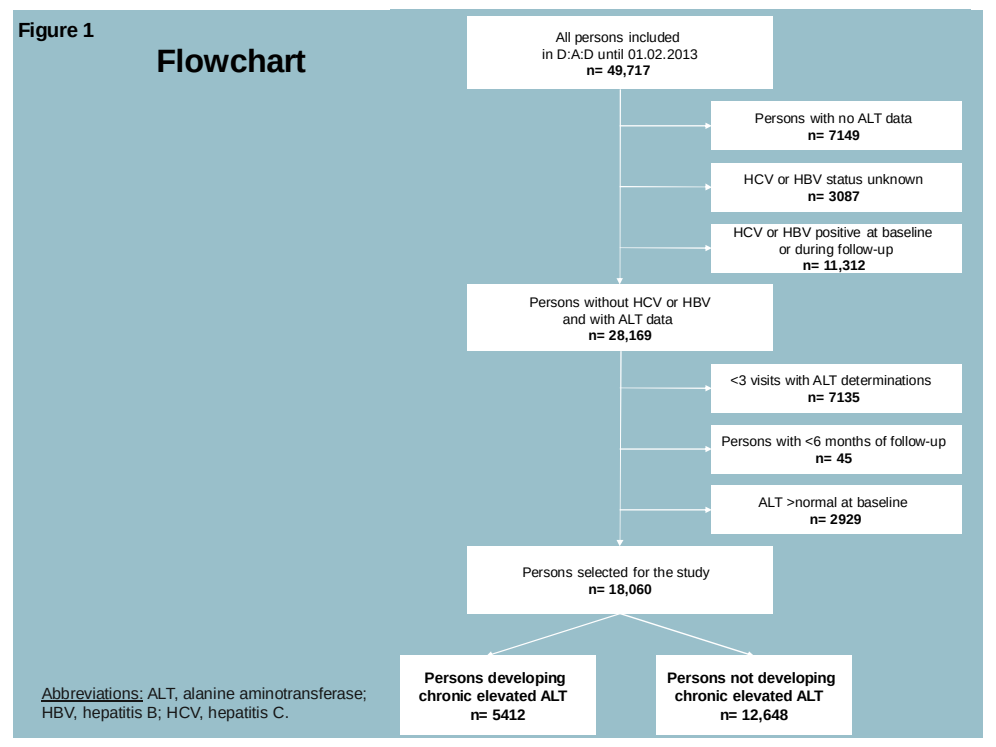

Flowchart
All persons included in D:A:D until 01.02.2013 n= 49,717
Persons with no ALT data n= 7149
HCV or HBV status unknown n= 3087
HCV or HBV positive at baseline or during follow-up n= 11,312
Persons without HCV or HBV and with ALT data n= 28,169
<3 visits with ALT determinations n= 7135
Persons with <6 months of follow-up n= 45
ALT >normal at baseline n= 2929
Persons selected for the study n= 18,060
Persons developing chronic elevated ALT n= 5412
Persons not developing chronic elevated ALT n= 12,648

Abbreviations: ALT, alanine aminotransferase; HBV, hepatitis B; HCV, hepatitis C.

Table 1 **Characteristics of 18,060 D:A:D study participants without HCV or HBV co-infection at study entry**

Total of participants, n (%)		18,060 (100%)
Sex, n (%)	Male	12,923 (71.6)
Age, years	median (IQR)	39 (33-48)
Ethnicity, n (%)	White	11,086 (61.4)
	Black	2095 (11.6)
	Other/unknown	4879 (27.0)
Mode of HIV transmission, n (%)	heterosexual	7740 (42.9)
	homosexual	8854 (49.0)
	injection drug use	313 (1.7)
	other/unknown	1153 (6.4)
Duration of D:A:D cohort follow-up, years	median (IQR)	7.1 (4.2-10.7)
Previous clinical AIDS	n (%)	4103 (22.7)
CD4 cells/µL	median (IQR)	449 (298- 630)
HIV-1 RNA ≤50 copies/mL	n (%)	7861 (43.5)
Ever received antiretroviral therapy	n (%)	13,406 (74.2)
Ever received NRTIs	n (%)	13,193 (73.1)
Ever received PIs	n (%)	10,008 (55.4)
Ever received NNRTIs	n (%)	7395 (41.0)
Body mass index, kg/m², n (%)	≤26	12,646 (70.0)
	>26	3343 (18.5)
Diabetes mellitus	n (%)	563 (3.1)

Abbreviations: ALT, alanine aminotransferase; HDL, high density lipoprotein; HBV, hepatitis B virus; HCV, hepatitis C virus; IQR, interquartile range; NNRTI, nonnucleoside reverse transcriptase inhibitor; NRTIs, nucleoside reverse transcriptase inhibitor; PI, protease inhibitor.

Table 2 **Uni- and multivariable Poisson regression models for demographic, anthropometric and clinical factors associated with chronic ALT elevations**

		Univariable Analysis		Multivariable Analysis*	
		RH (95% C.I.)	P-value	RH (95% C.I.)	P-value
Calendar year	2010-2013	1 (Ref.)	-	1 (Ref.)	-
	1999-2001	1.69 (1.53-1.86)	<0.001	1.89 (1.69-2.10)	<0.001
Sex	Female	1 (Ref.)	-	1 (Ref.)	-
	Male	0.96 (0.90-1.02)	0.16	0.88 (0.83-0.94)	<0.001
Age, years	<29	1 (Ref.)	-	1 (Ref.)	-
	≥ 60	0.74 (0.65-0.85)	<0.001	0.58 (0.50-0.67)	<0.001
Ethnicity	White	1 (Ref.)	-	1 (Ref.)	-
	Black	0.84 (0.77-0.91)	<0.001	0.84 (0.76-0.93)	<0.001
	Other	1.28 (1.12-1.47)	<0.001	1.24 (1.08-1.42)	0.002
	Unknown	0.95 (0.88-1.02)	0.15	1.02 (0.82-1.27)	0.86
Total cholesterol, mmol/L	< 6.2	1 (Ref.)	-	1 (Ref.)	-
	≥ 6.2	1.27 (1.19-1.35)	<0.001	1.57 (1.41-1.75)	<0.001
Triglycerides, mmol/L	<2.3	1 (Ref.)	-	1 (Ref.)	-
	≥ 2.3	1.48 (1.40-1.56)	<0.001	1.13 (1.02-1.25)	0.03
Lipodystrophy	No	1 (Ref.)	-	1 (Ref.)	-
	Yes	1.12 (1.06-1.18)	<0.001	1.41 (1.25-1.58)	<0.001
Body mass index, kg/m²	≥18, ≤26	1 (Ref.)	-	1 (Ref.)	-
	<18	0.79 (0.66-0.96)	<0.001	0.81 (0.67-0.98)	0.03
	>26, ≤30	1.30 (1.21-1.39)	<0.001	1.33 (1.24-1.43)	<0.001
	>30	1.28 (1.15-1.42)	<0.001	1.33 (1.19-1.49)	<0.001
Arterial hypertension	No	1 (Ref.)	-	1 (Ref.)	-
	Yes	1.06 (0.99-1.13)	0.09	1.11 (1.03-1.19)	0.006
On ART		1 (Ref.)	-	1 (Ref.)	-
Not on ART	VL <5 log10 c/mL	0.91 (0.84-0.98)	0.009	0.95 (0.88-1.03)	0.18
	VL ≥5 log10 c/mL	1.25 (1.04-1.49)	0.02	1.30 (1.08-1.56)	0.005

*adjusted for use of lipid lowering drugs, smoking and cohort.
Abbreviations: ALT, alanine aminotransferase; ART, antiretroviral therapy; LEE, liver enzyme elevation; VL, viral load.

- In the multivariable analysis, earlier years (1999-2001 vs 2010-2013), younger age, increased body mass index, dyslipidemia, use of lipid-lowering drugs, arterial hypertension and high HIV RNA levels were associated with chronic LEE. Black ethnicity and male gender were inversely correlated (**Table 2**).
- Chronic LEE was associated with ongoing exposure to regimens containing ddI, d4T, TDF, FTC, EFV and NVP. No evidence for an association with increased risk was found for 3TC, ABC, AZT, and all tested PIs (**Table 3**). EFV exposure for ≥2 years was inversely correlated.
- Because the association with TDF was unexpected, we further analysed commonly used TDF-containing regimens (**Figure 2**). The association was found to be more pronounced when TDF was used in combination with FTC and/or EFV.

Sensitivity analyses

- Using a LEE definition of a single ALT value >100 U/L, the observed association with TDF remained unchanged.
- There was no evidence that the TDF effect was modified by analyzing exclusively ART-naïve persons initiating ART.
- When using a common definition of chronic ALT elevation (>50 U/L) for both men and women, the gender effect reversed.

CONCLUSION

- Whilst ddI, d4T, NVP and EFV have been described to be hepatotoxic [3-4], we observed an additional association between TDF and chronic LEE emerging within first 2 years after drug initiation.
- The TDF signal seems to be enhanced when TDF is used in the combination with FTC and/or EFV.
- The results are consistent with other small case studies [5-8].
- The reasons for and clinical implications of this novel TDF-LEE signal calls for further investigations.

Table 3 **Uni- and multivariable Poisson regression models for categorical ART exposure associated with chronic ALT elevations**

		Univariable Analysis		Multivariable Analysis*	
		RR (95% C.I.)	P-value	RR (95% C.I.)	P-value
ddI	No exposure	1 (Ref.)	-	1 (Ref.)	-
	Currently on, exposed for <2years	1.75 (1.53-1.99)	<0.001	1.26 (1.08-1.45)	0.002
	Currently on, exposed for ≥2years	1.36 (1.24-1.51)	<0.001	1.27 (1.14-1.43)	<0.001
d4T	No exposure	1 (Ref.)	-	1 (Ref.)	-
	Currently on, exposed for <2years	1.90 (1.63-2.21)	<0.001	1.51 (1.26-1.82)	<0.001
	Currently on, exposed for ≥2years	1.50 (1.36-1.65)	<0.001	1.17 (1.04-1.33)	0.01
3TC	No exposure	1 (Ref.)	-	1 (Ref.)	-
	Currently on, exposed for <2years	1.03 (0.92-1.16)	0.56	0.88 (0.75-1.02)	0.09
	Currently on, exposed for ≥2years	0.98 (0.91-1.06)	0.26	0.98 (0.87-1.11)	0.78
FTC	No exposure	1 (Ref.)	-	1 (Ref.)	-
	Currently on, exposed for <2years	1.28 (1.18-1.39)	<0.001	1.17 (1.03-1.32)	0.02
	Currently on, exposed for ≥2years	0.80 (0.73-0.89)	<0.001	1.01 (0.86-1.18)	0.93
TDF	No exposure	1 (Ref.)	-	1 (Ref.)	-
	Currently on, exposed for <2years	1.46 (1.36-1.57)	<0.001	1.57 (1.41-1.75)	<0.001
	Currently on, exposed for ≥2years	0.88 (0.81-0.95)	0.001	1.17 (1.04-1.33)	0.01
EFV	No exposure	1 (Ref.)	-	1 (Ref.)	-
	Currently on, exposed for <2years	1.64 (1.50.1.79)	<0.001	1.13 (1.02-1.25	0.03
	Currently on, exposed for ≥2years	0.92 (0.85-1.00)	0.05	0.81 (0.73-0.90)	<0.001
NVP	No exposure	1 (Ref.)	-	1 (Ref.)	-
	Currently on, exposed for <2years	1.94 (1.74-2.16)	<0.001	1.41 (1.25-1.58)	<0.001
	Currently on, exposed for ≥2years	1.12 (1.02-1.23)	0.02	1.01 (0.91-1.13)	0.84
LPV	No exposure	1 (Ref.)	-	1 (Ref.)	-
	Currently on, exposed for <2years	0.76 (0.67-0.86)	<0.001	0.81 (0.67-0.97)	0.03

*Adjusted for exposure to the other antiretrovirals, age, sex, race, body mass index, lipids, use of lipid lowering drugs, lipodystrophy, hypertension, smoking, year, cohort and for each drug discontinued for <2 years and ≥2 years.

Figure 2

Incidence rate ratios and 95% CIs for the development of chronic ALT elevation

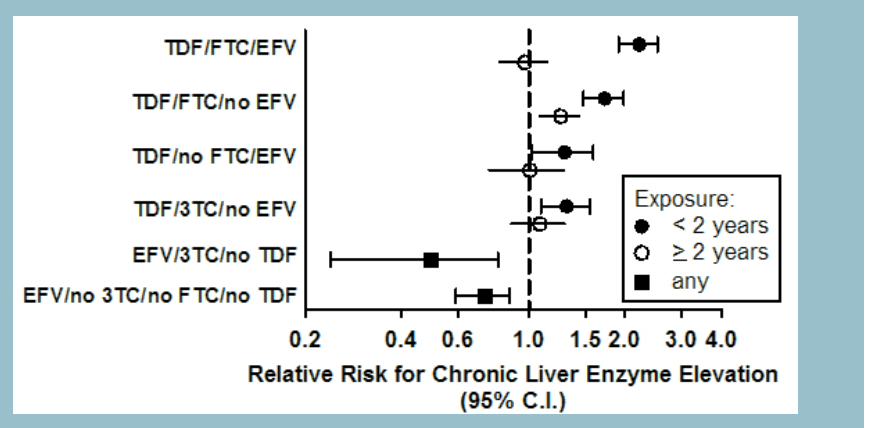

TDF/FTC/EFV
TDF/FTC/no EFV
TDF/no FTC/EFV
TDF/3TC/no EFV
EFV/3TC/no TDF
EFV/no 3TC/no FTC/no TDF
Exposure:
< 2 years
≥ 2 years
any
0.2 0.4 0.6 1.0 1.5 2.0 3.0 4.0
Relative Risk for Chronic Liver Enzyme Elevation (95% C.I.)

From Poisson regression models adjusted for exposure to the other antiretrovirals, age, sex, race, BMI, lipids, use of lipid lowering drugs, lipodystrophy, hypertension, smoking, year and cohort.

References

[1] Pol S et al. Clin Infect Dis 2004. [2] Bansi L et al. J Acquir Immune Defic Syndr 2009. [3] Kovari H et al. Clin Infect Dis 2010. [4] Ryom L et al. Poster No. 787; CROI 2014. [5] Lattuada E et al. AIDS 2008. [6] Qayyum S et al. Curr Drug Saf 2012. [7] Fink DL et al. Int J STD AIDS 2013. [8] Echenique IA. AIDS Patient Care STDS 2013.

Acknowledgements

Steering Committee: Members indicated w/ *; ¢ chair;
Cohort PIs: W El-Sadr* (CPCRA), G Calvo* (BASS), F Dabis* (Aquitaine), O Kirk* (EuroSIDA), M Law* (AHOD), A d'Arminio Monforte* (ICONA), L Morfeldt* (HivBIVUS), C Pradier* (Nice), P Reiss* (ATHENA), R Weber* (SHCS), S De Wit* (Brussels)

Cohort coordinators and data managers: M Hillebreght, S Zaheri, L Gras (ATHENA), M Bruyand, S Geffard, E Pernot, J Mourali (Aquitaine), H McManus, S Wright (AHOD), S Mateu, F Torres (BASS), M Delforge (Brussels), G Bartsch, G Thompsen (CPCRA), J Kjær, D Kristensen (EuroSIDA), I Fanti (ICONA), E Fontas, K Dollet, C Caissotti (Nice), A Sundström, G Thulin (HivBIVUS), M Rickenbach (SHCS)

Statisticians: CA Sabin*, AN Phillips*, DA Kamara, CJ Smith, A Mocroft

D:A:D coordinating office: L Ryom, CI Hatleberg, RS Brandt, D Raben, C Matthews, A Bojesen, J Nielsen, JD Lundgren*¢

Member of the D:A:D Oversight Committee: B Powderly*, N Shortman*, C Moecklinghoff*, G Reilly*, X Franquet*

D:A:D working group experts: Kidney: L Ryom, A Mocroft, O Kirk*, P Reiss*, M Ross, CA Fux, P Morlat, O Moranne, AM Kesselring, DA Kamara, CJ Smith, JD Lundgren *¢. Mortality: CJ Smith, L Ryom, AN Phillips*, R Weber*, P Morlat, C Pradier*, P Reiss*, N Friis-Møller, J Kowalska, JD Lundgren*¢. Cancer: CA Sabin*, L Ryom, M Law*, A d'Arminio Monforte*, F Dabis*, M Bruyand, P Reiss*, CJ Smith, DA Kamara, M Bower, G Fätkenheuer, A Donald, A Grulich, JD Lundgren*¢

External endpoint reviewer: A Sjøl (CVD), P Meidahl (oncology), JS Iversen (nephrology)

Funding: 'Oversight Committee for The Evaluation of Metabolic Complications of HAART' with representatives from academia, patient community, FDA, EMA and a consortium of AbbVie, Boehringer Ingelheim, Bristol-Myers Squibb, Gilead Sciences, ViiV Healthcare, Merck, Pfizer, F. Hoffmann-La Roche and Janssen Pharmaceuticals

Download poster at: www.cphiv.dk

chip Centre for Health & Infectious Disease Research

Rigshospitalet

Faculty of Health and Medical Sciences, University of Copenhagen

UniversitätsSpital Zürich