CROI 2005, Poster #866

Changes over Time in the Use of Antiretroviral Therapy and Risk Factors for Cardiovascular Disease in the D:A:D Study

Caroline Sabin[1], Linda Morfeldt[2], Nina Friis-Møller[3], Martin Rickenbach[4], Peter Reiss[5], Antonella D'Arminio Monforte[6], Christian Pradier[7], Ole Kirk[8], Gonzalo Calvo[9], Matthew Law[10], Patrick Mercié[11], Wafaa El-Sadr[12], Stephane de Wit[13], Jens D Lundgren[3], on behalf of the D:A:D Study Group

[1]Royal Free and UC Medical School, London, UK; [2]HivBivus, Karolinska Hospital, Stockholm, Sweden; [3]Copenhagen HIV Programme (CHIP), Hvidovre University Hospital, Copenhagen, Denmark; [4]SHCS, Centre Hospitalier Universitaire Vaudois, Lausanne, Switzerland; [5]ATHENA, Academic Medical Center, University of Amsterdam, The Netherlands; [6]ICONA, L Sacco Hospital, University of Milan, Italy; [7]Nice Cohort, CHU Nice Hôpital de l'Archet, Nice, France; [8]EuroSIDA, Hvidovre University Hospital, Copenhagen, Denmark; [9]BASS, Autonomous University of Barcelona, Barcelona, Spain; [10]AHOD, National Centre in HIV Epidemiology and Clinical Research, Sydney, Australia; [11]Aquitaine Cohort, Bordeaux University Hospital, INSERM U593, Bordeaux, France; [12]CPCRA, Columbia University/Harlem Hospital, New York, NY, USA; [13]Saint-Pierre Cohort, C.H.U. Saint-Pierre Hospital, Brussels, Belgium;

Professor Caroline Sabin,
Department of Primary Care and Population Sciences,
Royal Free and UC Medical School, Rowland Hill Street,
London, UK, NW3 2PF
Tel: +44 207 830 2239 ext. 4752
Fax: +44 207 794 1224
E-mail: c.sabin@pcps.ucl.ac.uk

INTRODUCTION

- Over the last five years there has been growing recognition that a relationship exists between increased exposure to combination antiretroviral therapy (cART) and the incidence of cardiovascular disease (CVD).
- Furthermore, the reduction in mortality in those infected with HIV following the widespread use of cART has meant that infected individuals are now starting to experience clinical events typical of an aging population.
- These factors may have encouraged patients to make behavioural and lifestyle changes to reduce their CVD risk factor profile, and may have also influenced the choice of drugs making up the cART regimen.

AIMS OF STUDY

To describe changes over time in the risk factors for CVD and the type of cART received in the D:A:D Study, a large international multi-cohort study that includes information on 23,441 HIV+ve individuals from Europe, the US and Australia.[1]

METHODS

- Calendar time was split into four intervals (2000, 2001, 2002 and 2003) and patients under follow-up were classified according to their risk factor status and antiretroviral treatment at the end of each interval.
- The following risk factors for CVD were considered:
 - Current smoker
 - Presence of hypertension
 - Total cholesterol [TC] >=6.2 mmol/L *or* HDL-cholesterol (HDL-c) <=0.9 mmol/L *or* TC:HDL-c ratio >=6.5*
 - Family history of CVD
 - Age >=45 in men/55 in women

**if one or more present then counted as one risk factor*

- For the purposes of this study, high-risk individuals were defined as those with a previous CVD event (prior MI, stroke or invasive procedure), diabetes or >=2 risk factors for CVD.
- Poisson regression methods were used to compare the incidence of MI in the four time periods before and after controlling for any changes in risk factors over time.

Reference

1. The Data Collection on Adverse Events of Anti-HIV Drugs (DAD) Study Group. Combination antiretroviral therapy and the risk of myocardial infarction. *N Engl J Med* 2004; **349**: 1993-2003.

Table 1

Frequency of risk factors for CVD over the period 2000-2003

	Calendar year			
	2000	2001	2002	2003
No. patients under follow-up	21494	22711	21465	20148
Male (%)	75.4	75.9	75.8	75.8
Aged ≥45 (men)/55 (women) (%)	24.9	27.7	30.5	34.3
Current smokers (%)	47.4	49.1	51.9	45.0
Ex-smokers (%)	16.0	16.4	17.3	24.7
Previous CVD (%)	1.1	1.4	1.6	1.7
Diabetes (%)	3.5	4.2	4.7	5.4
Receiving lipid-lowering drugs (%)	3.5	4.7	6.8	7.6
Lipodystrophy (%)	23.2	24.6	27.6	29.9
≥2 risk factors for CVD (%)	34.3	37.4	41.0	41.5
High-risk patients (%)	36.0	39.3	43.1	43.7

Figure 1

Box-plots showing (i) total cholesterol, (ii) HDL-cholesterol and (iii) triglycerides 2000-2003

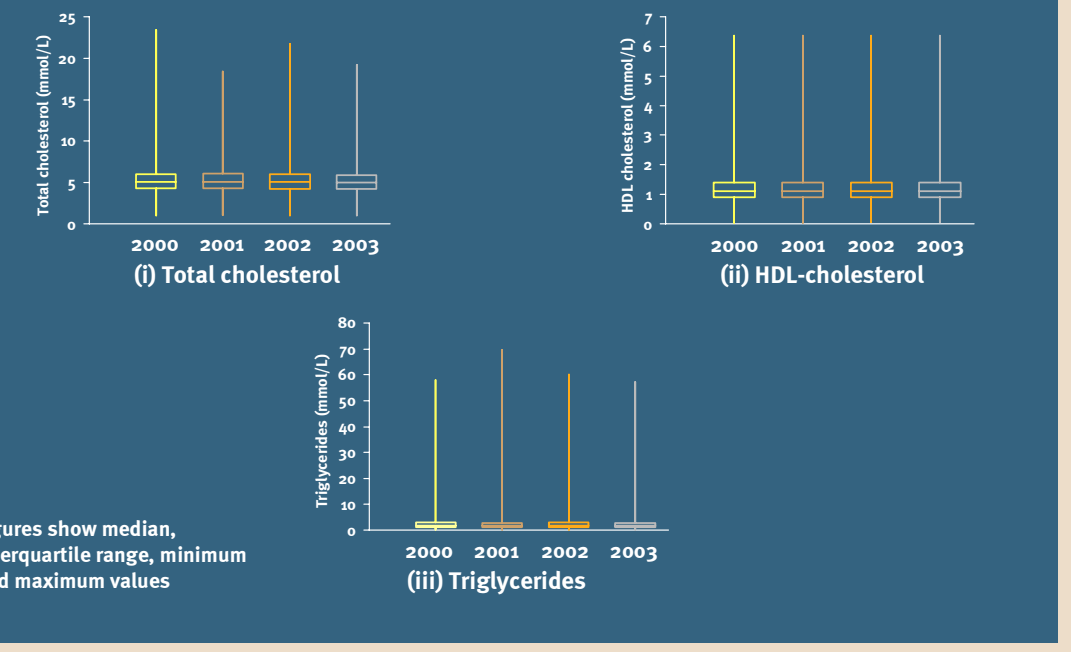
Total cholesterol (mmol/L)
0 5 10 15 20 25
2000 2001 2002 2003
(i) Total cholesterol
HDL cholesterol (mmol/L)
0 1 2 3 4 5 6 7
2000 2001 2002 2003
(ii) HDL-cholesterol
Triglycerides (mmol/L)
0 10 20 30 40 50 60 70 80
2000 2001 2002 2003
(iii) Triglycerides

Figures show median, interquartile range, minimum and maximum values

Figure 2

Pie charts showing drugs used as part of treatment regimens from 2000-2003

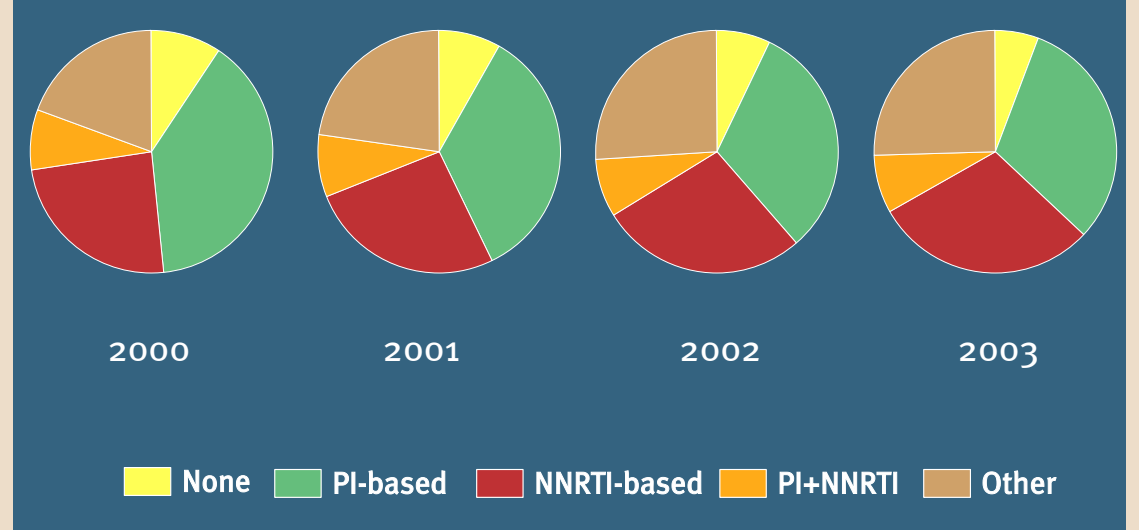
2000
2001
2002
2003
None
PI-based
NNRTI-based
PI+NNRTI
Other

RESULTS

- Changes in the prevalence of risk factors for CVD over the four-year period are shown in Table 1. As expected, the average age of those under follow-up increased as did the proportion of individuals with a previous CV event, the proportion with diabetes, the proportion receiving lipid-lowering drugs and the proportion with lipodystrophy. The proportion of individuals who were current smoker decreased in 2003 with a concomitant increase in the proportion of ex-smokers.
- Lipid levels remained relatively constant over time (Figure 1).
- The proportion of individuals with >=2 risk factors for CVD increased from 34.3% in 2000 to 41.5% in 2003; the proportion of patients deemed to be at high-risk of CVD increased from 36.0% to 43.7% over the same period.
- Changes in risk factors were generally more pronounced in men and those who were older, but were apparent in all groups. In particular, the proportions of patients with >=2 risk factors and who were at high-risk of CVD increased with time in both men and women and in those who were younger/older (Figure 3).
- At the same time, interventions also appear to be targeted to the persons at the highest risk, with a higher proportion of ex-smokers and of patients receiving lipid-lowering therapy in 2003 (Table 2).
- Although there was no univariable effect of calendar time on MI incidence (Table 3), adjustment for changes in CVD risk factors suggested that the rate of MI – all else being equal – was lower in later years compared to 1999/2000. However, MI risk continues to increase with longer exposure to cART (abstract #N-186).

CONCLUSIONS

- The CVD risk factor profile of patients in D:A:D has worsened over time, with an increase in the proportion of patients at high risk of CVD.
- After controlling for these changes, and for increased exposure to cART, the risk of MI has decreased over the years, possibly as a result of improved targeting of interventions to those at high risk (e.g. use of lipid-lowering drugs) or because of changes in the choice of cART.

Acknowledgements

Cohort principal investigators: G Calvo*, F Dabis*, W El Sadr* O Kirk*, M Law*, A d'Arminio Monforte*, L Morfeldt*, C Pradier*, P Reiss*, R Weber*, S De Wit*
Cohort coordinators and data managers: S Zaheri, I van Valkengoed, R Thiébaut, E Balestre, K Petoumenos, S Mateu, F Torres, B Sommereijns, B Poll, G Thompsen, J Kjær, P Pezzotti, E Fontas, C Caissotti, A Sundström, G Thulin, M Rickenbach, O Keiser
Statisticians: CA Sabin, AN Phillips *
Community representative: S Collins *
D:A:D coordinating office: N Friis-Møller, A Sawitz, JD Lundgren *¢
Steering Committee: Members indicated w/*; ¢ chair;
Additional members: E Loeliger *, T Mertenskoetter *, R Tressler *, I Weller *
Funding: 'Oversight Committee for The Evaluation of Metabolic Complications of HAART' with representatives from academia, patient community, FDA, EMEA and a consortium of 'Abbott, Agouron, Boehringer Ingelheim, Bristol-Myers Squibb, Gilead Sciences, GlaxoSmithKline, Merck, Pfizer, and Hoffman-La Roche'

Figure 3

Changes over time in the proportion of patients with >=2 risk factors for CVD and high-risk patients, stratified by (i) sex and (ii) age

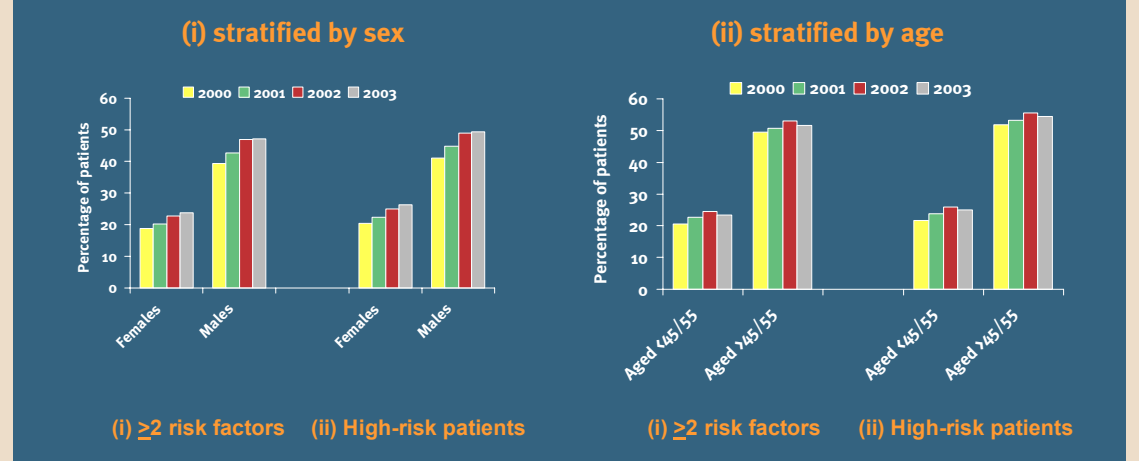
(i) stratified by sex
2000 2001 2002 2003
Percentage of patients
0 10 20 30 40 50 60
Females
Males
Females
Males
(i) ≥2 risk factors
(ii) High-risk patients
(ii) stratified by age
2000 2001 2002 2003
Percentage of patients
0 10 20 30 40 50 60
Aged <45/55
Aged >45/55
Aged <45/55
Aged >45/55
(i) ≥2 risk factors
(ii) High-risk patients

Table 2

Risk factors in individuals aged >45 (men)/55 (women) years, 2000-2003

	Calendar year			
	2000	2001	2002	2003
n	10169	11910	12441	12874
Male	83.4	83.1	82.3	81.6
Current smoker	42.8	45.6	49.0	42.3
Ex-smoker	18.6	18.8	19.8	27.1
Previous CVD	2.0	2.3	2.4	2.4
Diabetes	5.9	6.4	6.7	7.2
Previous stroke	0.9	1.0	1.1	1.1
TC (mmol/L)	5.4 (4.5, 6.4)	5.4 (4.5, 6.3)	5.3 (4.4, 6.2)	5.2 (4.4, 6.1)
HDL-c (mmol/L)	1.1 (0.9, 1.4)	1.1 (0.9, 1.4)	1.1 (0.9, 1.4)	1.1 (0.9, 1.4)
TG (mmol/L)	2.0 (1.3, 3.2)	2.0 (1.3, 3.2)	2.0 (1.3, 3.2)	1.9 (1.2, 3.0)
Use of lipid-lowering drugs	5.4	6.9	9.4	9.9

Table 3

Relative rates of myocardial infarction according to calendar year before and after adjusting for changes in risk factors

	Before adjustment			After adjustment		
Calendar year	Relative Hazard	95% CI	P-value	Relative Hazard	95% CI	P-value
2000	1	-	-	1	-	-
2001	1.18	0.82-1.68	0.38	1.13	0.79-1.61	0.51
2002	0.73	0.49-1.09	0.12	0.67	0.45-1.00	0.05
2003/4	0.70	0.46-1.05	0.09	0.62	0.41-0.93	0.02